Erasmus MC
Universitair Medisch Centrum Rotterdam

Abstract #365114: Use of Merlin Assay to identify patients with a low-risk for SN metastasis in a prospective multicenter Dutch study of a primary melanoma gene-signature (CP-GEP model) to predict sentinel node status during COVID-19.

Stassen R[1], Mulder E[1], Veringa A[2], Mooyaart A[3], Dwarkasing J[4], Tempel D[4], van der Hage J[5], Lendfers S[5], Aarts M[6], Verhoef C[1], Francken AB[2], Grunhagen D[1]

Background/ Methods

- Shift of resources during COVID-19 pandemic
- Sentinel Lymph Node Biopsy (SLNB) required various resources
- Majority of SLNBs is negative (70%-85%)
- Tool for identification of patients at low risk for SN-metastasis available.
- Is implementation feasible?

Methods

- Multicenter; Expert melanoma centers in the Netherlands
- Patients with primary cutaneous melanoma (T1-T4)
- Prospective open-label single arm non-interventional
- Feasibility of clinical implementation (turn-around time)
 - Interval signed consent -> report results
 - Interval shipping material -> report results

Informed consent → **Shipping of tissue** → **Return of results**

Logistic implementation of the CP-GEP model into clinical practice is feasible.

- Results in line with previous studies
- Accurate identification of patients at low risk for SN metastasis

Cohort Characteristics

Male	110 (57.9%)
Age (median [IQR])	64 (52-72)
Breslow Thickness (median [IQR])	1.4 (1.0-2.4)
AJCC T-classification	
T1	52 (27%)
T2	82 (43%)
T3	39 (21%)
T4	17 (9%)
High-Risk	120 (63.1%)
Low-Risk	70 (36.9%)

Results

Logistic implementation

Feasibility	First 50 patients	Last 50 patients
Intervals		
Signing PIF - Shipment Skyline (Median [IQR])	7 (6-11)	8 (7-12)
Shipment Skyline - Return of Report (Median [IQR])	8 (7-11)	8 (6-8)
Complete Interval (Median [IQR])	15 (14-21)	16 (14-20)

Surgery

underwent Re-excision + SLNB	160 (84.2%)
No SLNB	
Anatomical location	11 (5.8%)
Not found on pre-op Scintigraphy	6 (3.2%)
Shared decision making	13 (6.8)

Test results

	SN Positive	SN Negative	Missing	Total
High risk CP-GEP	25	75	20	120
Low risk CP-GEP	3	57	10	70
	28	132	30	190

NPV (overall): 95% | PPV (overall): 25%

Future Directions for Research

- Further optimization of logistic process
 - Shortening of process chain
 - Or; on-site testing possibility
- Expansion of participating centers
- Implementation into clinical practice

[1]Dept. Surg. Onc. Erasmus MC Cancer Institute, Rotterdam, Netherlands; [2]Dept Surg. Onc. Isala Hospital, Zwolle, Netherlands; [3]Dept. Pathology Erasmus MC Cancer Institute, Rotterdam, Netherlands; [4]SkylineDx, Rotterdam, Netherlands; [5]Dept. Surg. Onc. Leiden University Medical Center (LUMC), Leiden, Netherlands; [6]Dept. Onc. Maastricht University Medical Center, Maastricht, Netherlands;
Contact information: r.stassen@erasmusmc.nl